CLEO: A Phase 2 Randomized Controlled Adjunctive Treatment Trial with CLE-100 Esketamine Tablet for Patients with Major Depressive Disorder and Inadequate Response to Antidepressants, During The COVID-19 Pandemic

Maurizio Fava[1], Oren Berkowitz[2], Sanjay J. Mathew[3], Michael E. Thase[4], Andrew A. Nierenberg[5], Dan V. Iosifescu[6], L. Alison McInnes[7], Cristina Cusin[8], Robert Berman[9], Eli Eyal[2], Ari Gershon[2], Esther Lukasiewicz-Hagai[2]

[1] Massachusetts General Hospital, MGH Clinical Trials Network and Institute and Harvard Medical School, [2] Clexio Biosciences, [3] Baylor College of Medicine and Michael E. Debakey VA Medical Center, Houston and The Menninger Clinic,
[4] Department of Psychiatry, Perelman School of Medicine, University of Pennsylvania and the Corporal Michael J. Crescenz VAMC, [5] Dauten Family Center for Bipolar Treatment Innovation Massachusetts General Hospital and Harvard Medical School,
[6] NYU School of Medicine and Nathan Kline Institute, [7] Osmind, [8] Massachusetts General Hospital and Harvard Medical School, [9] Yale University School of Medicine and RMB Biotech Services

Introduction

CLE-100 is an investigational, oral esketamine tablet formulated with abuse deterrent technology for daily use at home as an adjunctive treatment for patients suffering from Major Depressive Disorder (MDD) with an inadequate response to antidepressants (AD).

- Esketamine and S-norketamine are potent NMDA receptor antagonists and block the ion channel at PCP site
 - The NMDA receptor plays a significant role in the etiology of depression[1,2]
- CLE-100 aims to be a safe and effective adjunctive antidepressant treatment for MDD patients with inadequate response to previous antidepressants:
 - To address unmet medical need: ~3 million patients in the US with MDD fail treatment with at least 2 different antidepressants[3]
 - With rapid onset as compared to SSRIs/SNRIs or atypical antipsychotics
 - With a favorable tolerability profile
 - Convenient, at home administration

1. Rosenbaum SB, Gupta V, Palacios JL. Ketamine. [Updated 2021 Feb 19]. In: StatPearls [Internet]. Treasure Island (FL): StatPearls Publishing; 2021 Jan.
2. Abdallah CG, Sanacora G, Duman RS, Krystal JH. Pharmacol Ther. 2018:190;148-158.
3. Zhdanava, M., Pilon, D., Ghelerter, I., Chow, W., Joshi, K., Lefebvre, P., & Sheehan, J. J. (2021). The Journal of Clinical Psychiatry, 82(2).

Orally Administered Esketamine is Metabolized to 2 Active Metabolites with NMDAR-Dependent and NMDAR-Independent Antidepressant Activities

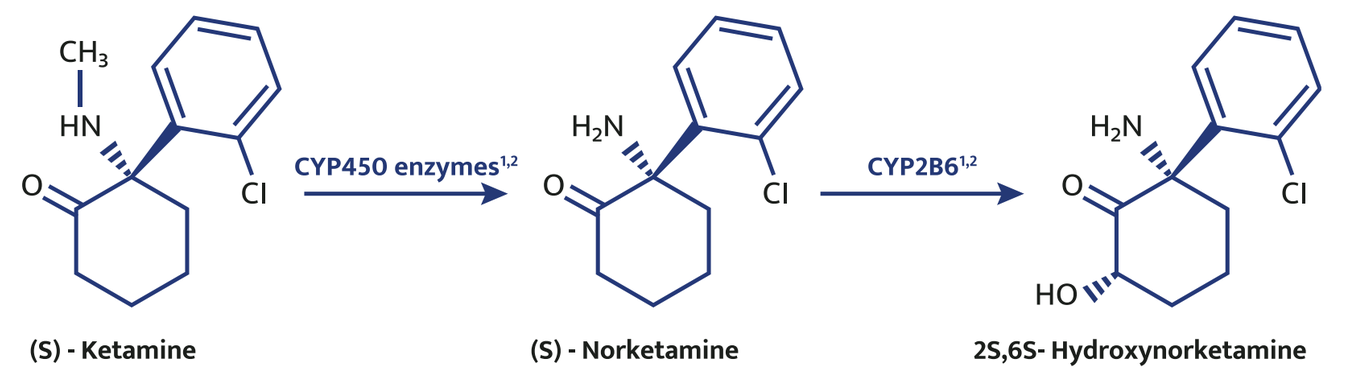

CYP450 enzymes[1,2]
CYP2B6[1,2]
(S) - Ketamine
(S) - Norketamine
2S,6S- Hydroxynorketamine

(S)-Ketamine[1]
- NMDAR antagonist[1]
- Demonstrated clinical antidepressant efficacy via multiple routes of administration in humans

(S)-Norketamine[2]
- NMDAR antagonist
- AMPA- and NMDAR-independent antidepressant effects in animals
- Favorable safety profile[2]

2S,6S-Hydroxynorketamine[3]
- Acute and sustained antidepressant effects in animals
- Mechanism of action appears to be independent of NMDAR

1. Molero P, et al. CNS Drugs. 2018;32(5):411-420. 2. Yang C, et al. Biol Psychiatry. 2018;84(8):591-600. 3. Yokoyama R, et al. Pharmacol Biochem Behav. 2020;191:172876

Methods

CLEO: A Phase 2 RCT Study

A 4 week study to assess the efficacy, safety and tolerability of daily at-home CLE-100 40 mg as adjunct treatment of MDD in patients with an inadequate response to ≥ 2 previous antidepressants

1:1 Randomization N=122
- CLE-100 (+ background AD) n=61
- Placebo (+ background AD) n=61

Screening (up to 28 days) → Double Blind Phase Daily Treatment(day 1 to day 29) → Follow Up (day 30 to day 42)

Primary Endpoint:
- Change in Montgomery–Åsberg Depression Rating Scale (MADRS) from baseline to week 4

Key Secondary Endpoints:
- Change in Clinician Global Impression – Severity (CGI-S) scale from baseline to week 4
- Change in Self report of Symptoms of Depression Questionnaire (SDQ) from baseline to week 4
- Change in Sheehan Disability Scale (SDS) from baseline to week 4
- Change in MADRS from baseline to week 2

CLEO: Key Entry Criteria

Key Inclusion Criteria
- Male or female 18 to 65 years old (inclusive)
- Primary diagnosis of MDD without psychotic features
- MADRS score of at least 24 at Screening verified by SAFER rater
- Inadequate response to at least 2 (up to 5) previous antidepressant therapies* in the current Major Depressive Episode (MDE) confirmed by SAFER rater
- Current MDE length: 12 weeks to 5 years

* Using the MGH-ATRQ (Massachusetts General Hospital Antidepressant Treatment Response Questionnaire)

Key Exclusion Criteria
- History of substance use disorder (except tobacco) within past 12 months or lifetime Hx of psychedelic use disorder
- Has had previous nonresponse for treatment of depression to ketamine or esketamine
- History or current diagnosis of Bipolar disorder, schizophrenia, or schizoaffective disorders within lifetime
- Other predominant mental disorders within 1 year of screening (e.g., PTSD, OCD)
- High risk of suicide
- SPB>140mmHg or DBP >90mmHg or any medical condition for which an increase in blood pressure or intracranial pressure poses a serious risk

CLEO: Statistical Analyses

- The primary endpoint (PEP) was change in MADRS score from baseline at Week 4.
- A mixed model repeated measures (MMRM) analysis was used to estimate the difference in the estimated Least Square Means (LSM) at week 4 between the CLE-100 and the placebo groups.
- The recruitment period spanned from August 2020 to August 2022, coinciding initially with the acute phase of the COVID-19 pandemic, followed by a post-acute phase beginning in 2022 that led to a return towards normal life. A post-hoc analysis was performed to compare the phases of the pandemic by adding them to the MMRM model.

Results

CLEO: Study Disposition

- The study randomized 130 subjects across 32 US sites; 125 subjects were included in the primary analysis
- Study completion: 92% in CLE-100 treatment group vs. 83% in placebo. In CLE-100 treatment group, no Early Termination due to AE
- Demographics and Baseline Depression characteristics were similar across both treatment groups

Demographics characteristics	CLE-100 40 mg (n=72)	Placebo (n=58)
Mean age (SD)	42.8 (13.2)	46.2 (13.1)
Gender (%)		
• Female	51 (71%)	43 (74%)
• Male	21 (29%)	15 (26%)
Race (%)		
• White	54 (75%)	46 (79%)
• Black/Afro-American	11 (15%)	10 (17%)
• Asian	6 (8%)	0%
• American Indian or Alaska native	0%	1 (2%)
• Other	1 (1%)	1 (2%)
Ethnicity (%)		
• Hispanic or Latino	15 (21%)	24 (41%)
• Not Hispanic not Latino	57 (79%)	33 (57%)
• Not reported		1 (2%)
Mean BMI (SD)	30 (5.2)	29.5 (4.4)

Baseline characteristics	CLE-100 40 mg (n=72)	Placebo (n=58)
Mean MADRS (SD)	32.9 (5.0)	32.9 (4.9)
AD response in current MDE		
• Non responders (%)	38 (53%)	36 (62%)
• Partial Responders (%)	34 (47%)	22 (38%)
• Inadequate response to 2 AD in current MDE (%)	58 (81%)	48 (83%)
• Inadequate response to > 2 AD in current MDE (%)	14 (19%)	10 (17%)
Mean current MDE Duration (days) (SD)	716 (476)	782 (483)
Mean age at first MDE (years) (SD)	24.4 (12.2)	25.6 (12.5)
Background AD (%)		
• SSRI	46 (64%)	29 (50%)
• SNRI	9 (13%)	14 (24%)
• Tricyclics	2 (3%)	0 (0%)
• Other antidepressants	17 (24%)	15 (26%)

CLEO Efficacy Results: PEP (MADRS W4 change from baseline)

	CLE-100 40 mg (n=70)	Placebo (n=55)
MADRS baseline, mean (SD)	32.9 (5.0)	33.0 (4.9)
Ls mean change (SE)	**-10.7 (1.1)**	**-9.5 (1.3)**
Ls mean difference from placebo (SE)	**-1.26 (1.7)**	
P-value	0.46	

PEP in the overall sample was not statistically significant: −1.26 [1.69], 95% CI: [-4.93 to 1.58], P-value = 0.46

CLEO Study Recruitment Spanned August 2020-2022

Acute Phase of Pandemic: March 2020 → December 2021
- Exceptional efforts invested to reduce transmission of the virus through lockdown, widespread testing, exhaustive contact tracing, country-wide vaccination campaigns
- CLEO: First-Patient-In: August 2020; 63 patients enrolled in 2020-2021, over 16 months

Post-acute Phase of Pandemic: January 2022 → August 2022
- December 2021: White House announces a new phase in the pandemic response: boost vaccinations to continue to fight this virus without shutting down our schools and businesses
- 2022: Transition towards a sustainable level of response focused on the management of severe outcomes and protecting vulnerable populations (i.e. de-escalating measures)
- CLEO: 67 patients enrolled in 2022, over 8 months; Last-Patient-In: August 2022

CLEO Post-hoc Efficacy Analysis: Temporal Impact on PEP

Change in MADRS from Baseline to Week 4 in Subjects enrolled in 2020-2021 (adjusted mean)

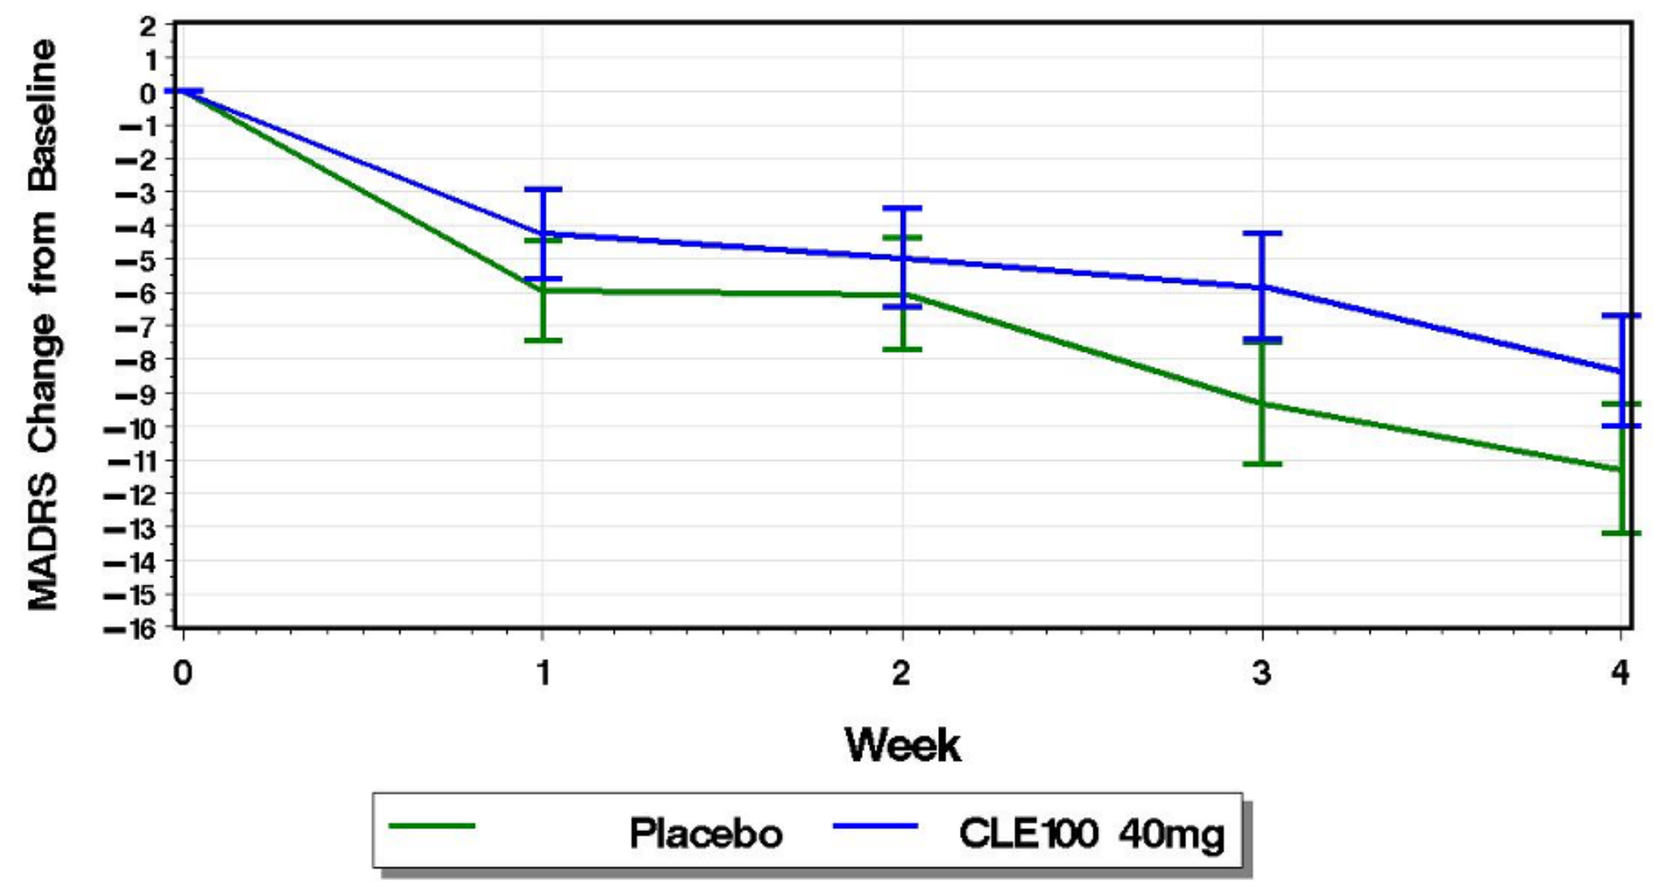

MADRS Change from Baseline
2
1
0
−1
−2
−3
−4
−5
−6
−7
−8
−9
−10
−11
−12
−13
−14
−15
−16
0
1
2
3
4
Week
Placebo
CLE100 40mg

During 2020-2021 (n=60):
- Placebo (n=25): -11.3 (1.9)
- CLE-100 (n=35): -8.3 (1.6)
- PEP difference from placebo: 2.9 (2.4), p=0.23

Change in MADRS from Baseline to Week 4 in Subjects enrolled in 2022 (adjusted mean)

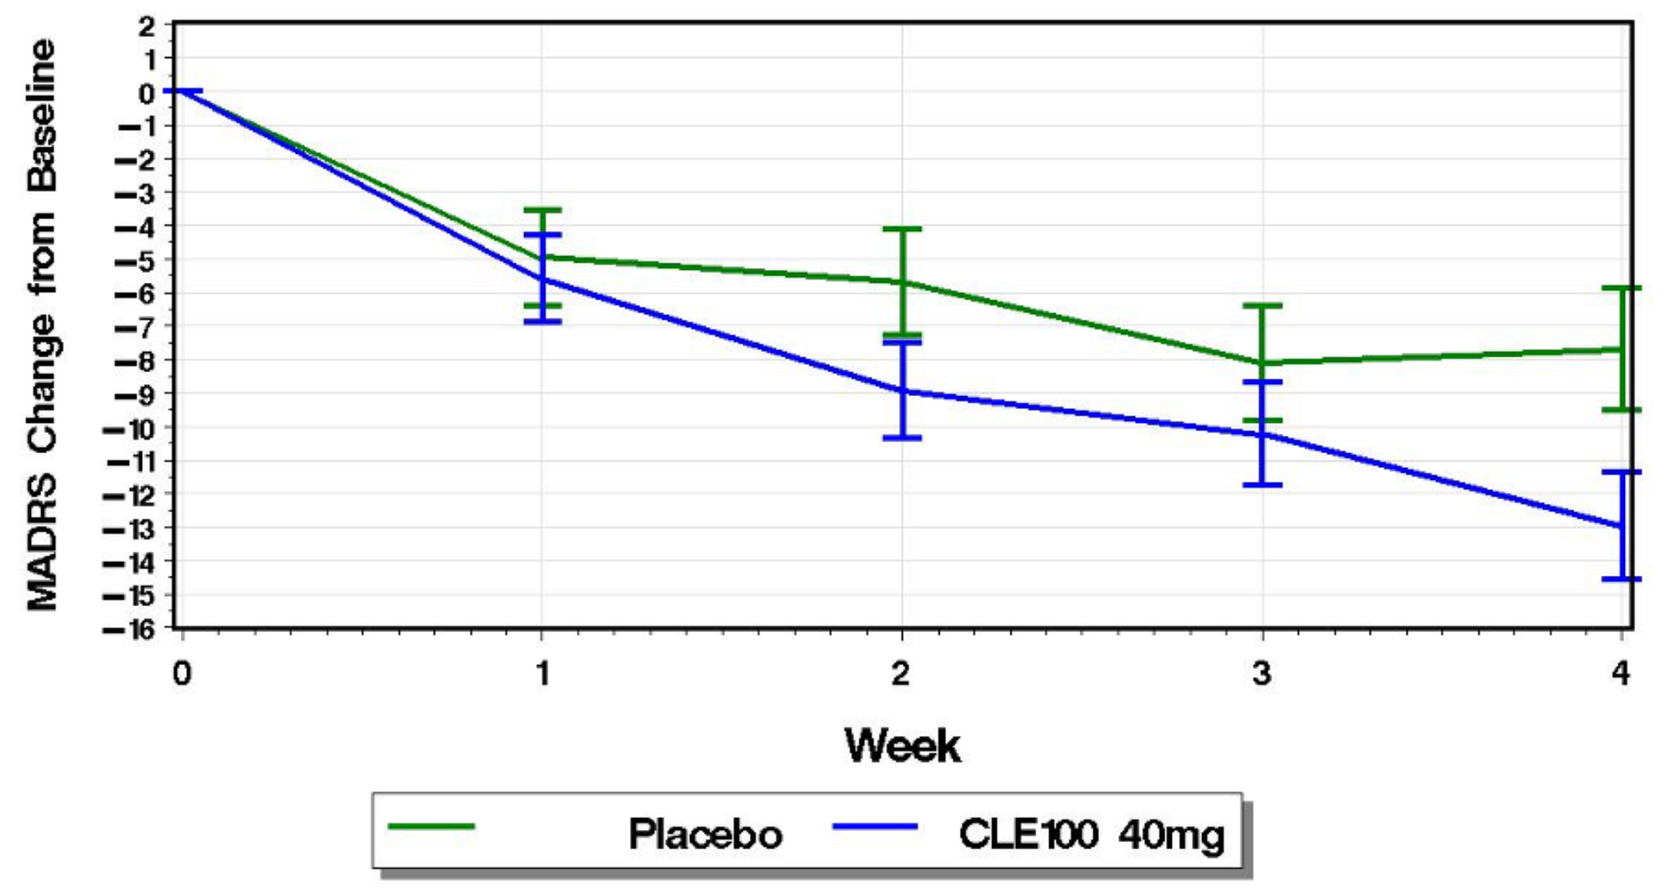

MADRS Change from Baseline
2
1
0
−1
−2
−3
−4
−5
−6
−7
−8
−9
−10
−11
−12
−13
−14
−15
−16
0
1
2
3
4
Week
Placebo
CLE100 40mg

In 2022 (n=65):
- Placebo (n=30): -7.7 (1.8)
- CLE-100 (n=35): -12.9 (1.6)
- **PEP difference from placebo: -5.3 (2.3) , p=0.026**
- CGI-S (key SEP): -0.7 (0.3) , p=0.04

CLEO: Safety and Tolerability Results

- Number (%) of subjects who had at least 1 TEAE: 37 (51.4%) in CLE-100, 23 (40.4%) in placebo
- No adverse events were serious in either treatment arm
- Number (%) of TEAEs that led to discontinuation of study medication: 0 (0.0%) in CLE-100, 5 (10.9%) in placebo
- In CLE-100 arm, most of the AEs were mild (78.1%) with only 2 severe AEs (both anxiety) in 2 subjects
- Very low rate of dissociation in both arms
- Drug abuse, dependence, or withdrawal were not observed

TEAEs occurring in more than 2% of CLE-100 subjects and more frequently than in the placebo	CLE-100 40 mg (n=72)			Placebo (n=57)		
	Events	N	% of N	Events	N	% of N
Dizziness	17	10	13.9 %	2	1	1.8 %
Headache	11	9	12.5 %	5	5	8.8 %
Somnolence	9	8	11.1 %	1	1	1.8 %
Nausea	10	6	8.3 %	0	0	0 %
Vomiting	4	3	4.2 %	0	0	0 %
Anxiety	4	3	4.2 %	0	0	0 %
Vision blurred	4	2	2.8 %	0	0	0 %
Upper respiratory tract infection	2	2	2.8 %	1	1	1.8 %
Nasopharyngitis	2	2	2.8 %	0	0	0 %
Dissociation	3	2	2.8 %	3	1	1.8 %
Dysuria	2	2	2.8 %	0	0	0 %

CLEO: Neuropsychiatric Safety Scales and Blood Pressure

- Potentially clinically significant Clinician-Administered Dissociative State Scale (CADSS) scores (> 4) occurred in 12.5% of CLE-100 and 10.5 % of placebo subjects
- For CLE-100: the maximum mean increase in CADSS from pre-dose was observed on Day 1 at 0.5 h: 0.68 points
- Post-dose sedation assessed by the Modified Observer's Alertness/Sedation scale (MOAA/S) was observed in 12 (16.7%) of CLE-100 subjects and 5 (8.8%) of placebo subjects
- Minimum worst post-dose MOAA/S score was 4* in both arms
- In the CLE-100 arm, sedation assessed by MOAA/S resolved in all CLE-100 subjects by 3hrs post-dose
- 4-Item Brief Psychiatric Rating Scale (BPRS): no meaningful differences between the two arms
- Suicidality: Subjects shifted from no suicidal ideation or behavior to suicidal ideation more frequently in the placebo arm (10.5% of subjects) than in the CLE-100 arm (5.6% of subjects)
- Withdrawal: no signal for a withdrawal syndrome on the 20-item Physician Withdrawal Checklist (PWC-20)
- Cognition: Digit Symbol Substitution Test (DSST) scores shows no meaningful differences in post-dose DSST between the CLE-100 and placebo groups.
- Some transient small post-dosing increases in SBP and DBP in the CLE-100 arm and resolved by 3 hours post-dose
- The maximum mean increase in SBP was of 4.58 mmHg at 0.5 hours post-dose on Day 1 (and 2.00 mmHg in placebo arm)

*lethargic response to name spoken in normal tone

Discussion

Potential Factors for the Different Efficacy Results in the 2 Cohorts

- **Disruptive effect of acute COVID-19 pandemic on patients**
 - Clinical sites were struggling to enroll patients during the acute COVID-19 pandemic period.
 - Patients volunteering for the study during the acute phase of the pandemic might have differed from usual study populations in MDD treatment trials.
 - Psychosocial stressors arising from the acute COVID-19 pandemic – e.g. isolation, fear of death, loss of work – potentially could have triggered or exacerbated the MDD episodes of subjects, and may have impacted the subject's response to drug and placebo
- **Other potential factors**
 - There was no difference in treatment effect in 2022 between sites that had already opened early on (2020-21) and sites opened later in the study (2021-22) (i.e. sites that enrolled during both periods)
 - No meaningful differences were found in baseline characteristics between the two cohorts (2020-21 vs. 2022)

Conclusions

- CLE-100 is a novel, investigational oral esketamine representing a mechanistically new approach for the treatment of depression
- CLEO did not meet its PEP in the overall study population.
- Post-hoc analysis of subjects recruited in 2022, during the post-acute phase of the COVID-19 pandemic, demonstrated a clinically meaningful and statistically significant effect of CLE-100 40 mg on the PEP
- Robustness of the 2022 cohort PEP results is supported by positive results on CGI-S, an important SEP
- We hypothesize that these temporal differences were due to the disruptive impact of the pandemic, and thus believe that CLEO's results present a clear signal for CLE-100's potential efficacy
- CLE-100 40 mg was well tolerated with no serious adverse events. The safety profile seen in CLEO was compatible with at-home dosing
- The efficacy signal seen in the post-acute pandemic cohort along with the overall favorable safety and tolerability profile observed in this study deserve further exploration
- Additional studies of CLE-100 are being planned to confirm the positive results observed in the post-acute pandemic phase

Disclosures, past 3 years: M. Fava has received research support from: Abbvie; Acadia Pharmaceuticals; Aditum Bio Management Company, LLC; Allergan, Alkermes, Inc.; Altimate Health Corporation; Alto Neuroscience, Inc.; Ancora Bio, Inc.; Angelini S.p.A; Aptinyx; Arbor Pharmaceuticals LLC; Avanir Pharmaceuticals Inc.; Axsome; Benckiser Pharmaceuticals, Inc.; BioClinica, Inc.; Biogen; BioHaven; BioShin Limited; Cambridge Science Corporation; Centrexion Therapeutics Corporation; Cerecor; Cybin IRL Limited; Damona Pharmaceuticals; EmbarkNeuro; Eliem Therapeutics LTD; Gate Neurosciences, Inc.; GenOmind, LLC; Gentelon, LLC; Gerbera Therapeutics, Inc.; Happify; Johnson & Johnson; Lundbeck Inc.; Marinus Pharmaceuticals; Medpace, Inc.; Methylation Sciences, Inc.; Millennium Pharmaceutics, Inc.; Minerva Neurosciences; Neuralstem; Neurocrine Biosciences, Inc.; NeuroRX Inc.; Novaremed; Novartis; Otsuka; Pfizer; Premiere Research International; Praxis Precision Medicines; Protagenic Therapeutics, Inc.; Relmada Therapeutics Inc.; Reckitt; Shenox Pharmaceuticals; Stanley Medical Research Institute (SMRI); Taisho; Takeda; University of Michigan; Vistagen; WinSanTor, Inc.; Xenon Pharmaceuticals, Inc.; National Institute of Drug Abuse (NIDA); National Institutes of Health (NIH); National Institute of Mental Health (NIMH); and PCORI. Dr. Fava has not done any personal consulting. Any consulting he has done has been on behalf of Massachusetts General Hospital, except for Revival Therapeutics and Sensorium Therapeutics, he has spoken/published for Global Medical Education, Inc. Mood Disorders Summit, November 2020 and has equity Holdings in : Compellis; Neuromity; Psy Therapeutics; Revival Therapeutics; Sensorium Therapeutics and Royalty/patent, other income: Patents for Sequential Parallel Comparison Design (SPCD), licensed by MGH to Pharmaceutical Product Development, LLC (PPD) (US_7840419, US_7647235, US_7983936, US_8145504, US_8145505); and patent application for a combination of Ketamine plus Scopolamine in Major Depressive Disorder (MDD), licensed by MGH to Biohaven. Patents for pharmacogenomics of Depression Treatment with Folate (US_9546401, US_9540691) and has Copyright for the MGH Cognitive & Physical Functioning Questionnaire (CPFQ), Sexual Functioning Inventory (SFI), Antidepressant Treatment Response Questionnaire (ATRQ), Discontinuation-Emergent Signs & Symptoms (DESS), Symptoms of Depression Questionnaire (SDQ), and SAFER; Belvior; Lippincott, Williams & Wilkins; Wolkers Kluwer; World Scientific Publishing Co. Pte.Ltd. **S. Mathew** is supported through the use of resources and facilities at the Michael E. Debakey VA Medical Center, Houston, Texas and receives support from The Menninger Clinic, consultant to Almatica Pharma, Axsome Therapeutics, Biohaven, BioXcel Therapeutics, Boehringer-Ingelheim, Clexio Biosciences, COMPASS Pathways, Delix Therapeutics, Douglas Pharmaceuticals, Eleusis, EMA Wellness, Engrail Therapeutics, Janssen, Levo Therapeutics, Perception Neurosciences, Praxis Precision Medicines, Neumora, Neurocrine, Relmada Therapeutics, Sage Therapeutics, Seelos Therapeutics, Signant Health, Sunovion, and XW Pharma and has received research support from Boehringer-Ingelheim, Merck, Neurocrine, and Sage Therapeutics. **M. Thase** has consulted for Axsome, Clexio Pharma, GH Therapeutics, Janssen Pharmaceuticals, Inc., H. Lundbeck A/S, Otsuka Pharmaceutical Co., Ltd., Pfizer Inc., Sage Pharmaceuticals, Seelos Pharmaceuticals, Sunovion Pharmaceuticals Inc.,Takeda Pharmaceutical Company Ltd. Has received grants from Acadia, Alkermes, AbbVie, AssureRx Health (now Myriad), Axsome, Intracellular, Janssen Pharmaceuticals, Inc., National Institute of Mental Health, Otsuka, Patient Centered Outcomes Research Institute, Takeda, has received royalties from American Psychiatric Press., Inc., Guilford Publications, Herald House, Kluver-Wolters, W.W. Norton & Co, Inc and his spouse is senior VP of Open Health, dealing with many companies. D. Iosifescu has consulted for Axsome, Boehringer Ingelheim, Clexio, Neumora, Relmada, Sage. **L.A. McInnes** works for Osmind and is on the CAB of Clexio Biosciences. C. Cusin has consulted for Perception, Clexio , Compass and received research grants from: Livanova, ATAI, American Suicide Prevention Foundation and NIMH. **R. Berman** is a shareholder in Biohaven Pharmaceuticals, Gilgamesh Pharma, Freedom Biosciences, Gate Therapeutics, Osmol Therapeutics, Hemoshear Therapeutics, Manifest Technologies, and MindImmune Therapeutics. **E. Lukasiewicz Hagai, O. Berkowitz, E. Eyal, A. Gershon** are employees of Clexio Biosciences. **A.A. Nierenberg** has consulted for Alkermes, Clexio, Ginger/Headspace Health, Janssen, Merck, Neuronetics, NeuroRx, Otsuka, Protagenics, SAGE, Sunovion, Unravel. Has received honoraria from Belvior, EISAI, Wiley Depression and Anxiety, Psychiatric Annals Slack Publications. Has received royalties from Guilford Publications, Up-to-Date Wolters Kluwer Health. Has been on Adjudication Committee for Novartis. Has served on Scientific Advisory Board for Altimate, Flow, Milken Center for Strategic Philanthropy, Myriad, 4M Therapeutics